From Libraries of Small Molecule Macrocycles to Drugs

Helmut Thomas[1], Jennifer Filbey[2]

[1]Tranzyme Pharma Inc., 3001 12th Ave North, Sherbrooke, QC, Canada, J1H 5N4
[2]Tranzyme Pharma Inc., 4819 Emperor Drive, Suite 400, Durham, NC, USA, 27703

Summary

- No target limitations
- Validated drug discovery technology
- Confirmed hit to clinical candidate in as rapid as 18 months
- Robust manufacturing processes for large batches at high yields
- Generated multiple clinical drug candidates
- Unencumbered intellectual property landscape

Small Macrocycles With Drug-Like Properties

A proprietary drug discovery strategy based upon the efficient construction of diverse libraries of novel macrocyclic compounds is presented. These conformationally-defined molecules in combination with high throughput screening have generated multiple potent and selective hits against a wide range of pharmacological targets, including GPCRs, protein-protein interactions, proteases and ion channels. The unique chemical and biological features of this strategy include ease of SAR development, accelerated lead-to-clinic optimization, favorable safety and ADME properties, straightforward scalability, and an unencumbered patent landscape. Among the case studies that will be described are the identification and optimization of modulators of critical GPCR targets in the gastrointestinal tract that have led to compounds with appropriate properties to be developed as pharmaceuticals. The most advanced of these clinical candidates has now reached Phase III trials.

Linear peptide ligands

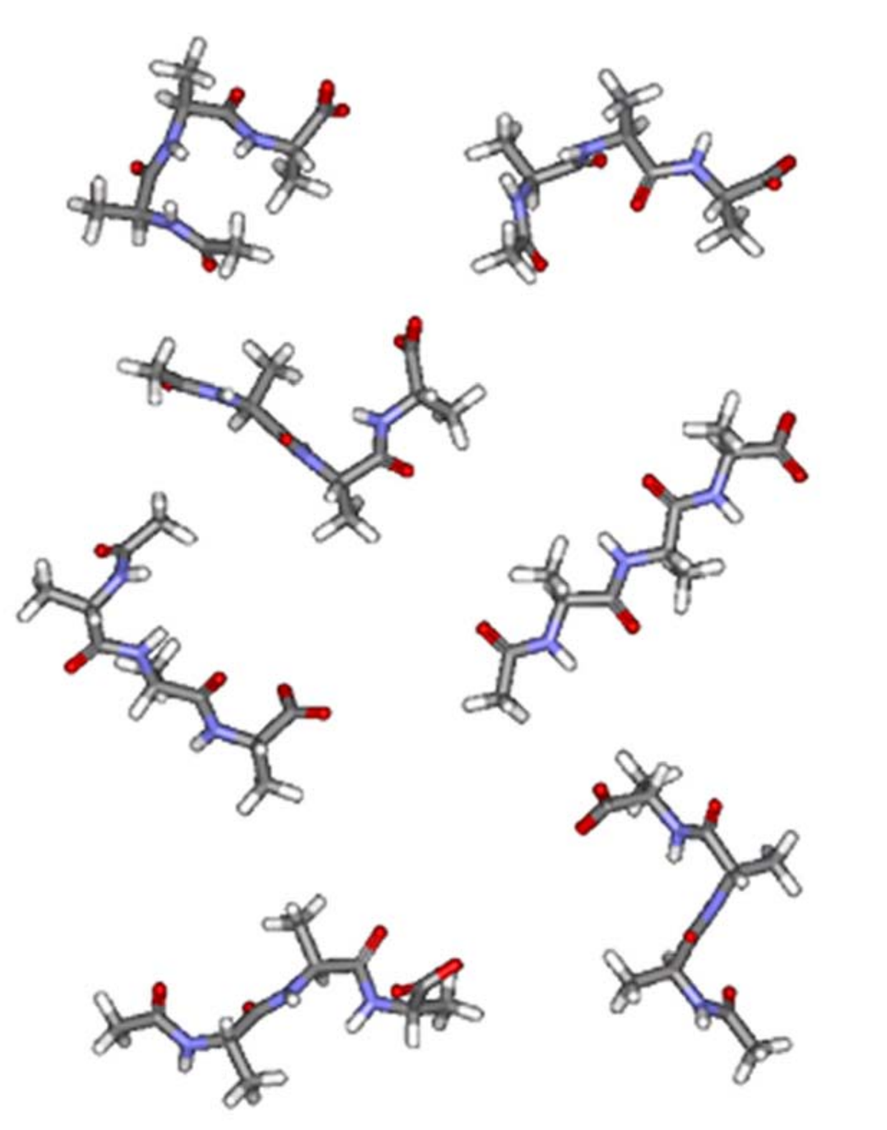

- Numerous uncontrolled conformers
- Substrate for proteases

Macrocyclize
Non-peptidic tether

Macrocyclic ligands

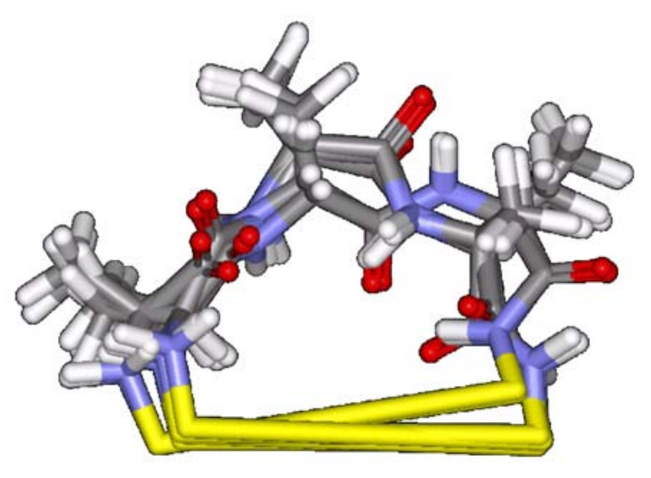

- Conformationally locked
- Reduced binding entropy / more potent
- High proteolytic stability
- Improved specificity and selectivity
- Solid SAR
- Recognized in drug discovery

Molecular Recognition Elements X, Y, Z
- Variable hydrophobicity / electrostatics
- Variable steric environment
- Variable orientation

Ionizable group
- Solubility
- Salt formation

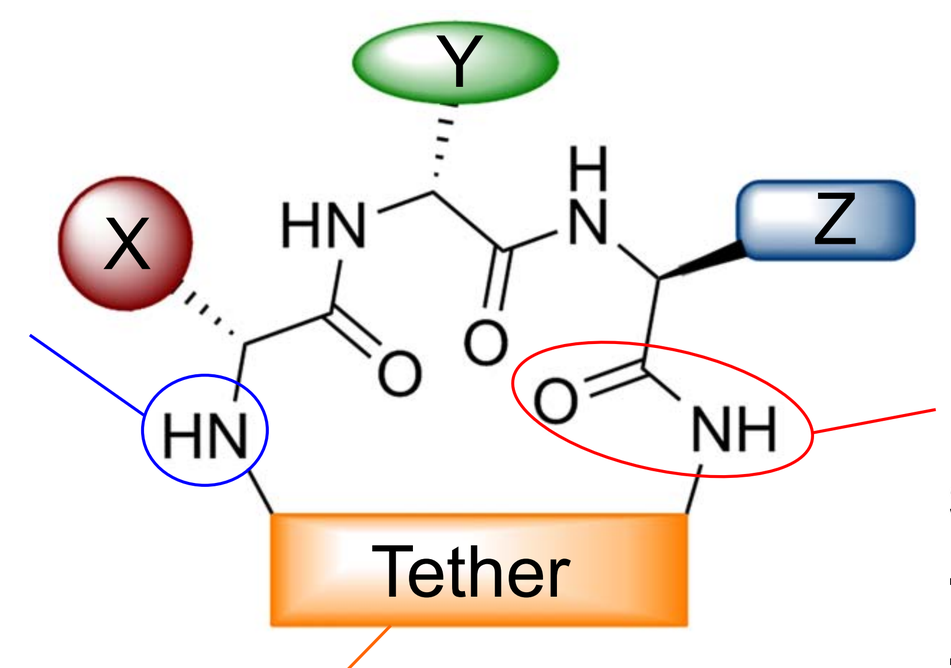
X
Y
Z
HN
H
N
O
HN
NH
Tether

Native amide bonds or peptide surrogates
- H-bond networking
- Chemical property modification

Tether
- Primary topological control
- Molecular recognition site
- PK/PD modulator

MATCH™ (Macrocyclic Template Chemistry) - Easily Scalable, Unique And Versatile Library Design

High Throughput Parallel Synthesis => Rapid Library Generation => Instant SAR

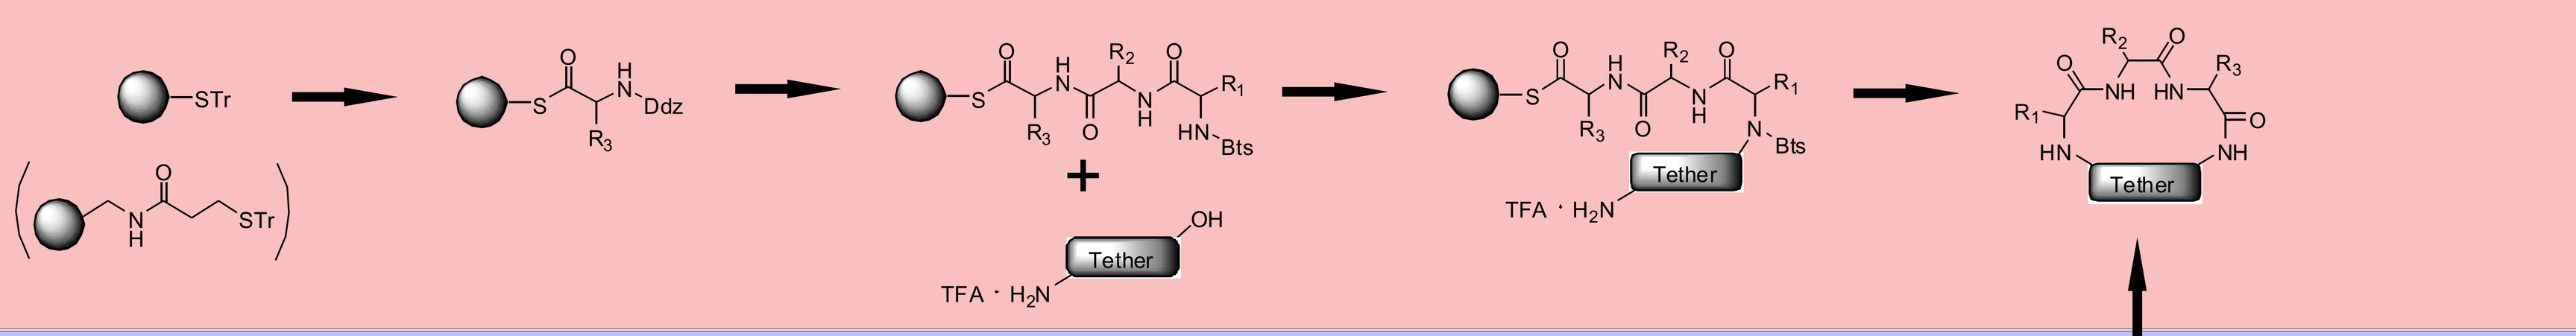
STr
Ddz
Bts
Tether
TFA · H2N
OH
NH
HN

Easily Scalable

- Convenient scaling from gram to multi-kg quantities
- Successfull manufacturing of multi-kg GMP batches
- Purification by crystallisation
- Very good yields

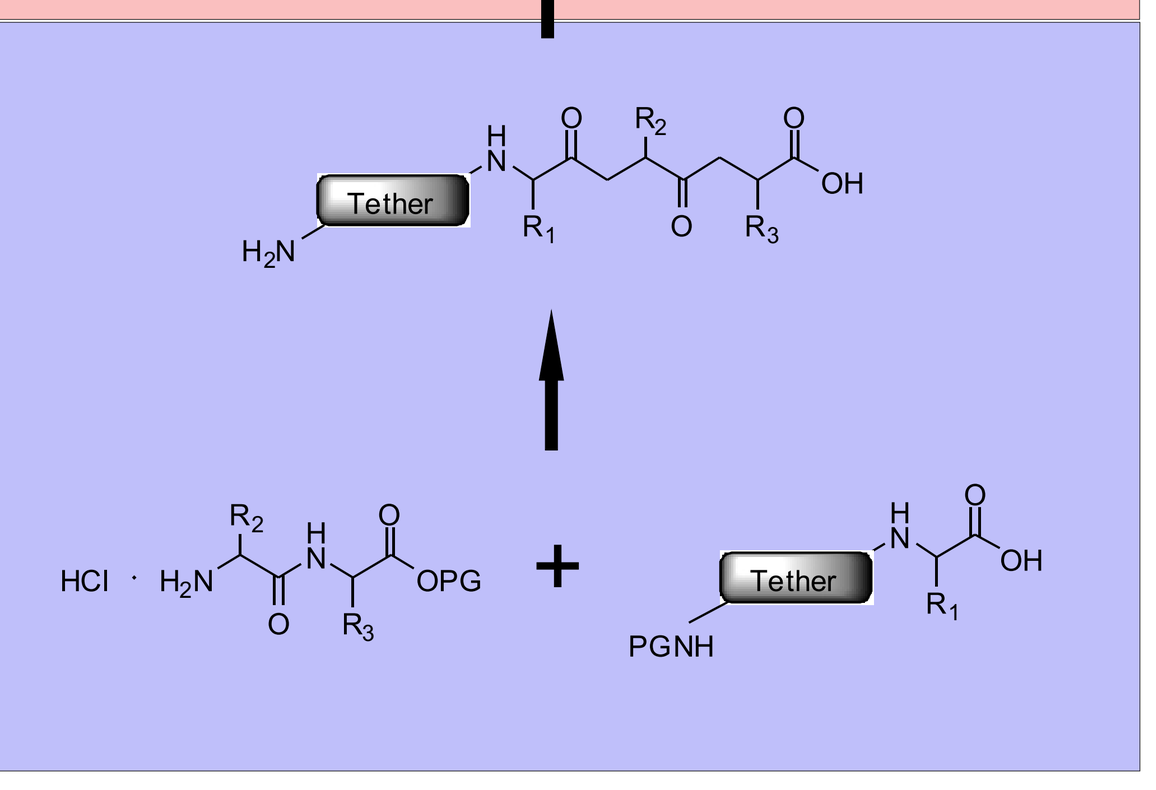
Tether
H2N
OH
HCl · H2N
OPG
PGNH

Compelling Reasons to Employ MATCH™ and HITCREATE™

- Unique small macrocycles (MWt ~500)
- Multiple recognition elements
- Tight topological control
- Unmatched diversity
- Ideal for rational design
- Straight-forward parallel synthesis
- High quality hit generation
- Accelerated, resource-efficient lead optimization
- Easily scalable chemistry
- Excellent chemical and metabolic stability
- Good oral bioavailability
- Superior safety profile

- No target limitations
- Successful clinical drug
- Strong IP position worldwide

- 40,000 compounds synthesized
- 20,000 compounds available for screening
- Hit rate of 0.2-0.5%
- High hit potentcy: initial K_i frequently < 500 nM
- Hit validation rate up to 50%
- High selectivity against liability targets (> 300 fold)
- Successfully applied to diverse target classes
 - GPCRs, ion channels, protein-protein, proteases

HITCREATE™ In Drug Discovery - HT Parallel Synthesis For Lead Optimization

Competitive Timelines from Hit to Clinical Candidate

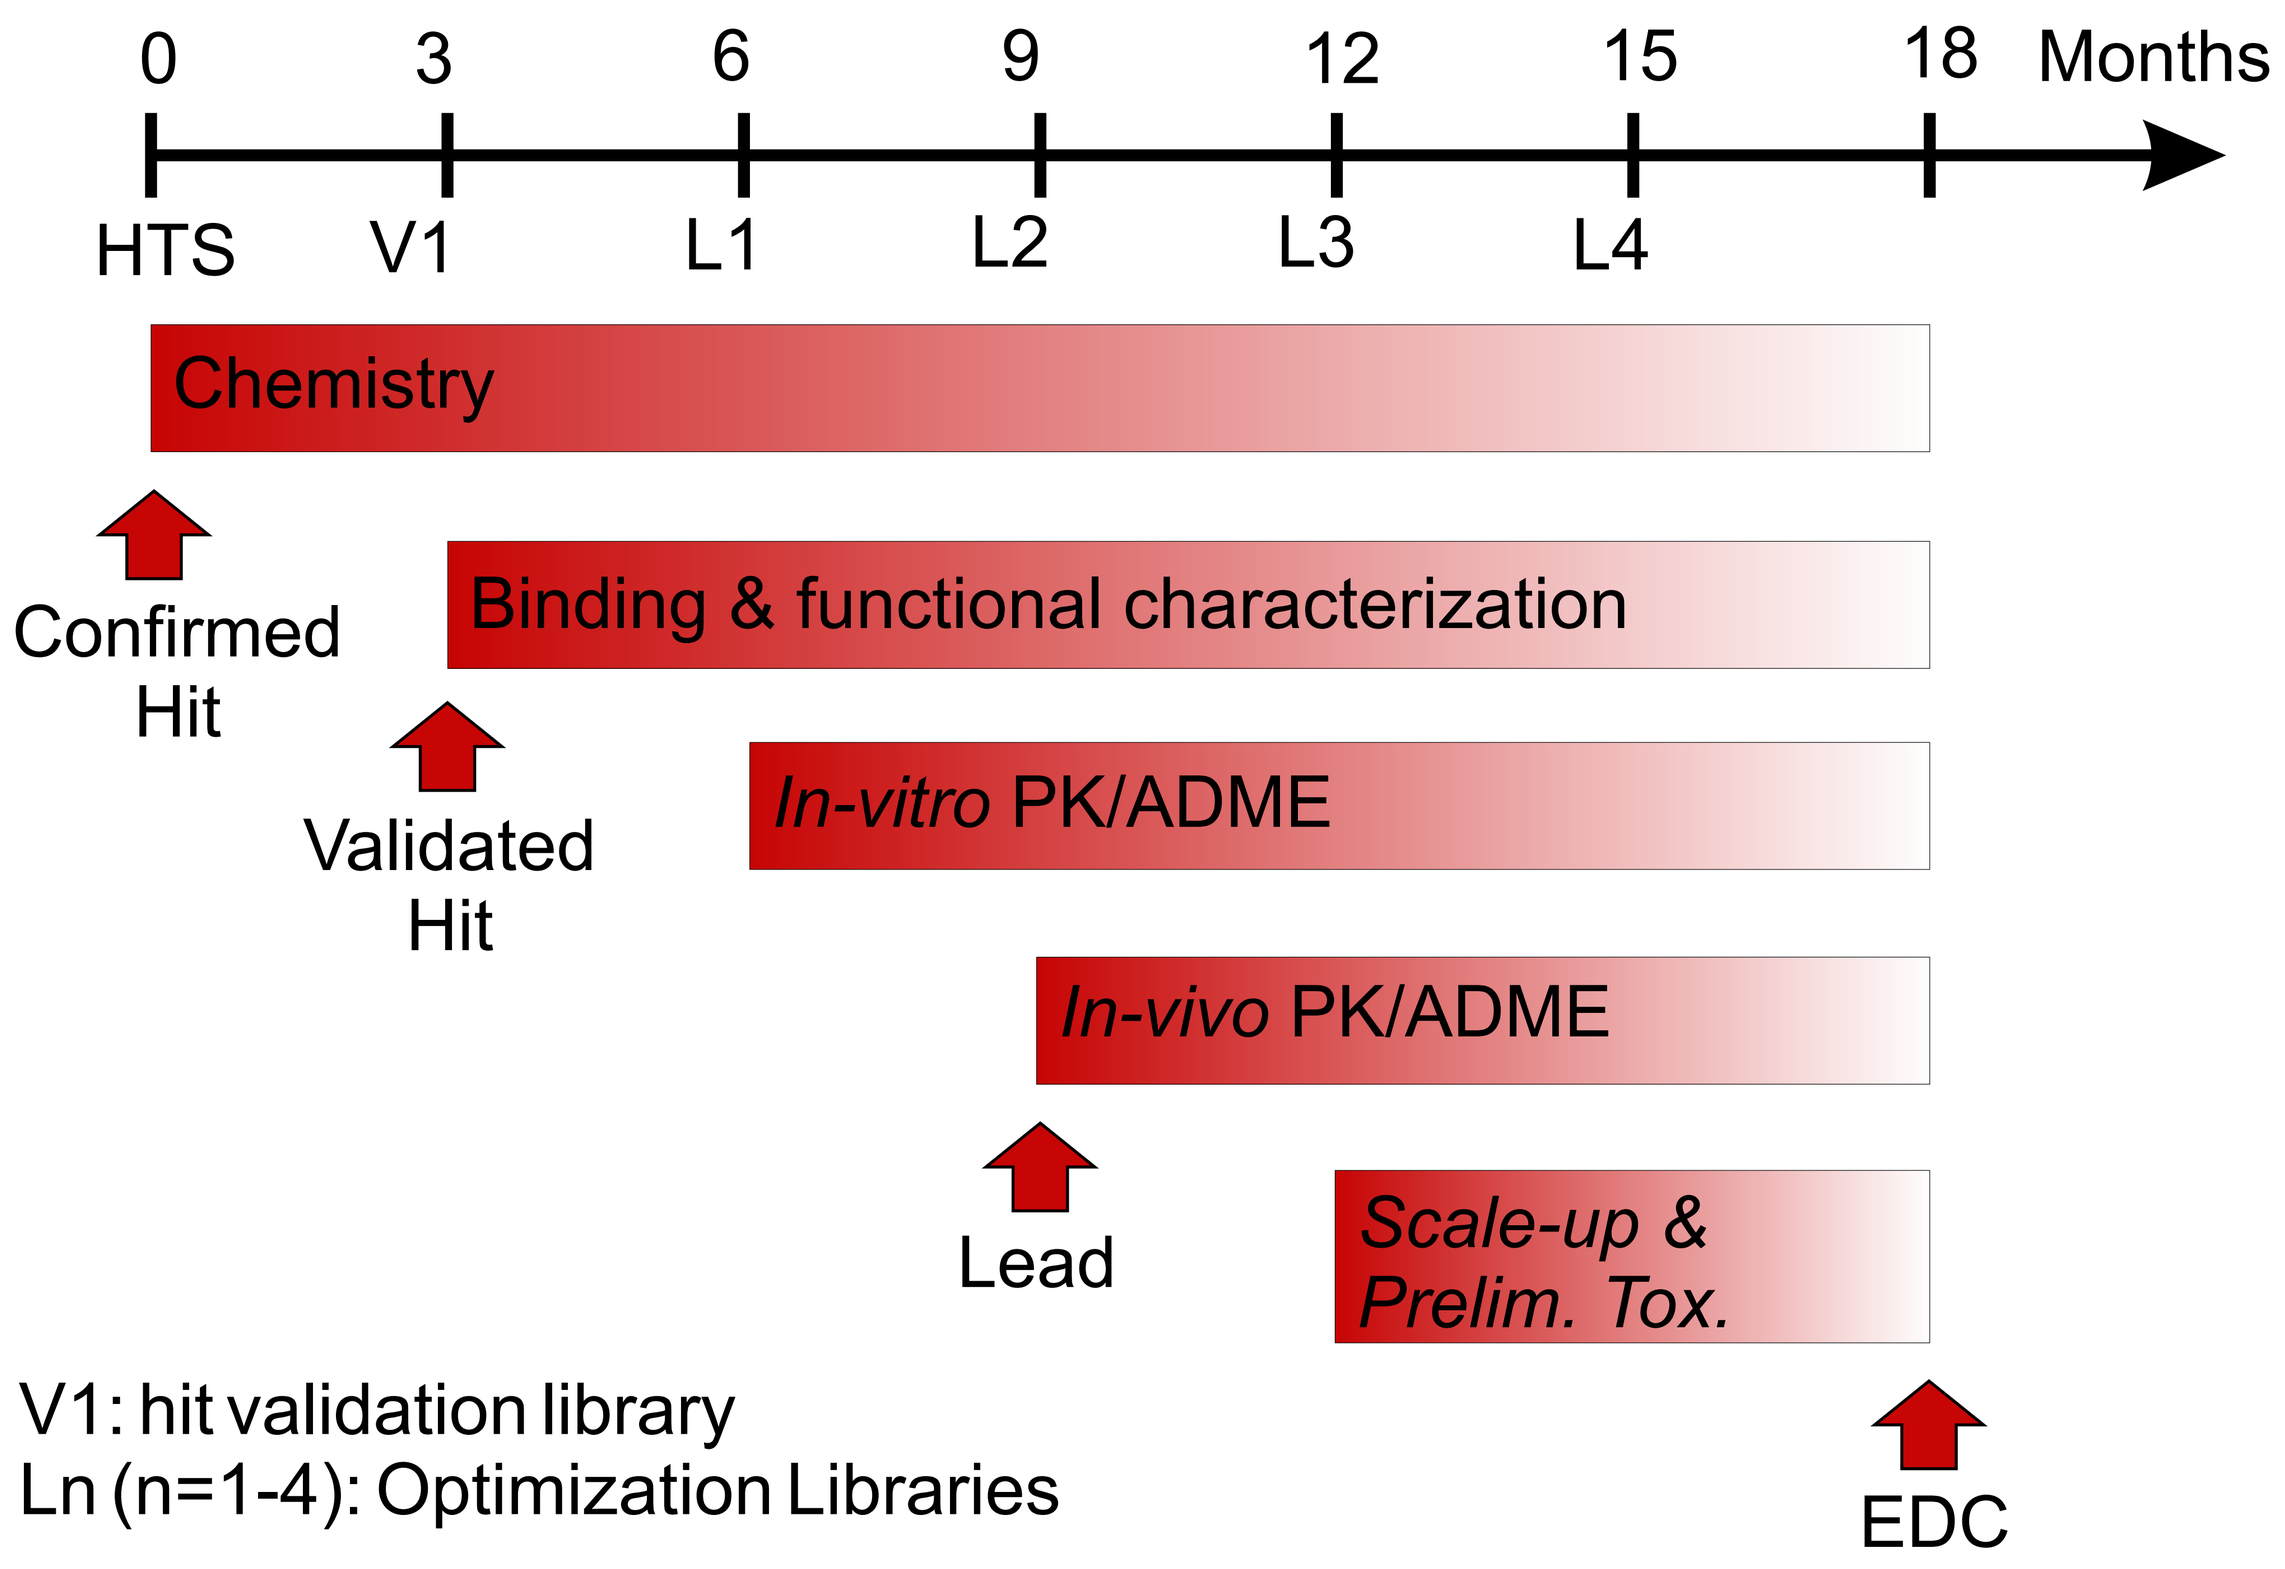
0
3
6
9
12
15
18
Months
HTS
V1
L1
L2
L3
L4
Chemistry
Confirmed Hit
Binding & functional characterization
Validated Hit
In-vitro PK/ADME
In-vivo PK/ADME
Lead
Scale-up & Prelim. Tox.
EDC

V1: hit validation library
Ln (n=1-4): Optimization Libraries

Representative Ghrelin Receptor Agonist Structures

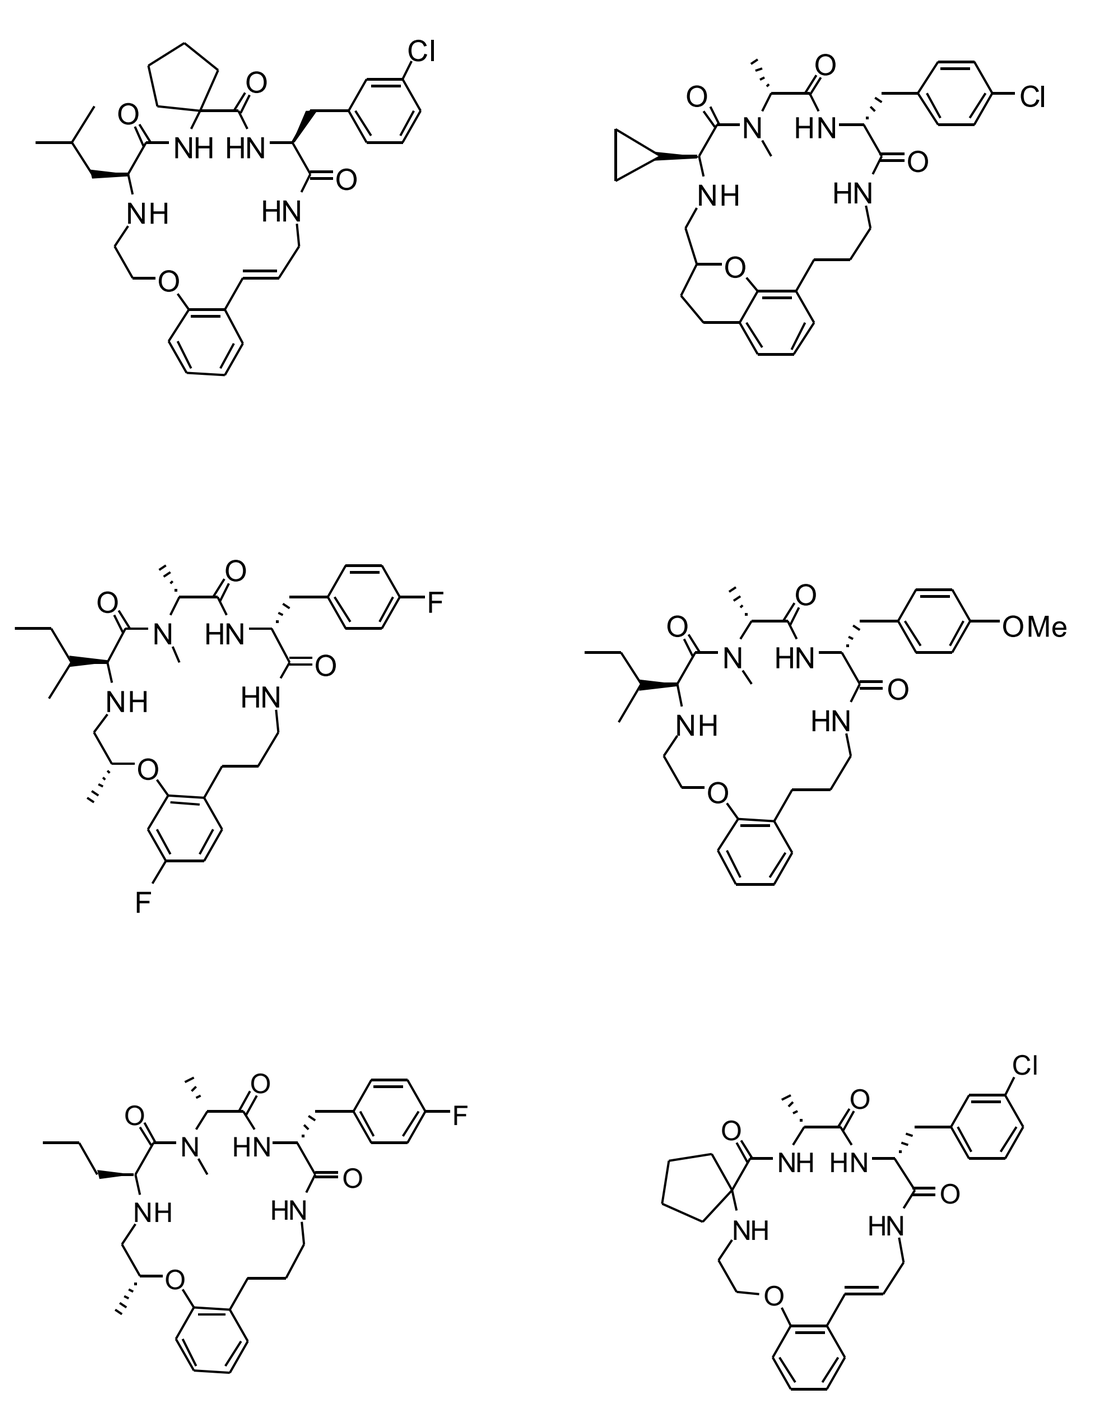

Key Properties of the Clinical Ghrelin

Property	TZP-101	TZP-102
Molecular Weight	~ 500	~ 500
ClogP	5.3	4.5
pK_a	7.17 ± 0.04	6.23 ± 0.11
K_i (hGRLN)	22 nM	12 nM
Ec_{50} (hGRLN)	27 nM	11 nM
ED_{GE} (rat, iv bolus)	0.1 mg/kg	ND
ED_{GE} (rat, oral)	>3 mg/kg	3 mg/kg
ED_{POI} (rat, iv bolus)	0.1 mg/kg	ND
IC_{50} CYP3A4	>0.5 μM (BFC/BQ)	>5 μM (BFC/BQ)
IC_{50} Other CYPs*	>50 μM	>100 μM
BBB Perm.	0.1 %	1.3 %
Oral BAV Rat	16 %	13 %
Dog	ND	25 %
Monkey	23 %	26 %
Man	ND	*ca* 85 %

* CYP2D6, 2C9, 2C8, 2C19, 2B6, 2A6, 1A2

MATCH™ Macrocycles Appear to Fit in the Active Site of Viral Proteases

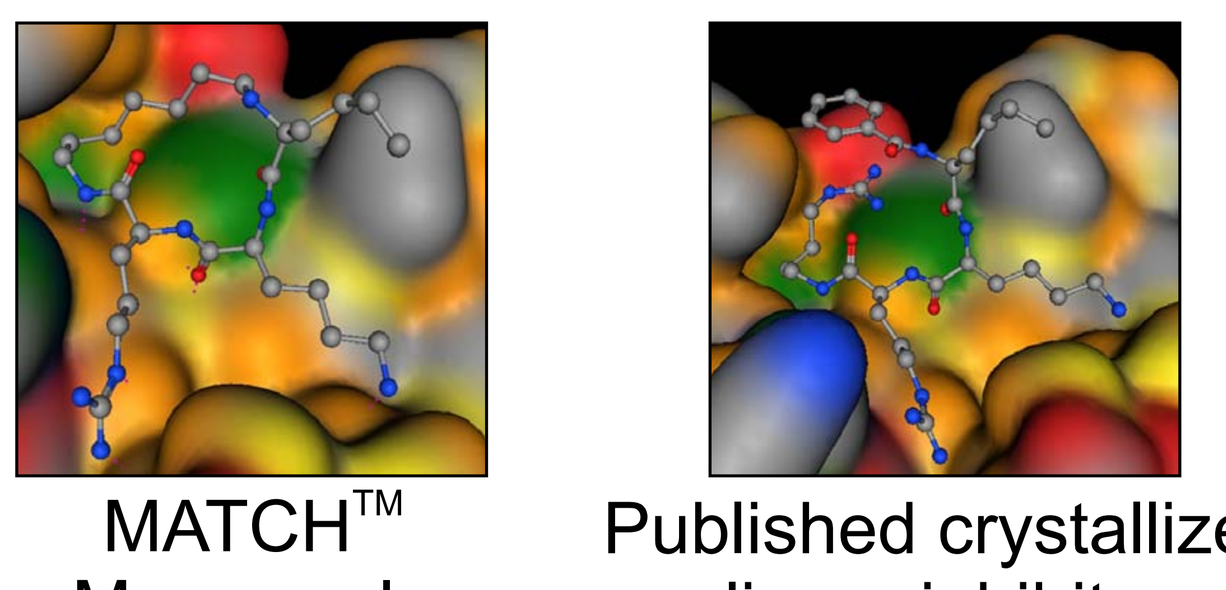

MATCH™ Macrocycle — Published crystallized linear inhibitor

A Potent and Selective Oncogenic Non-MMP Protease Inhibitor Hit

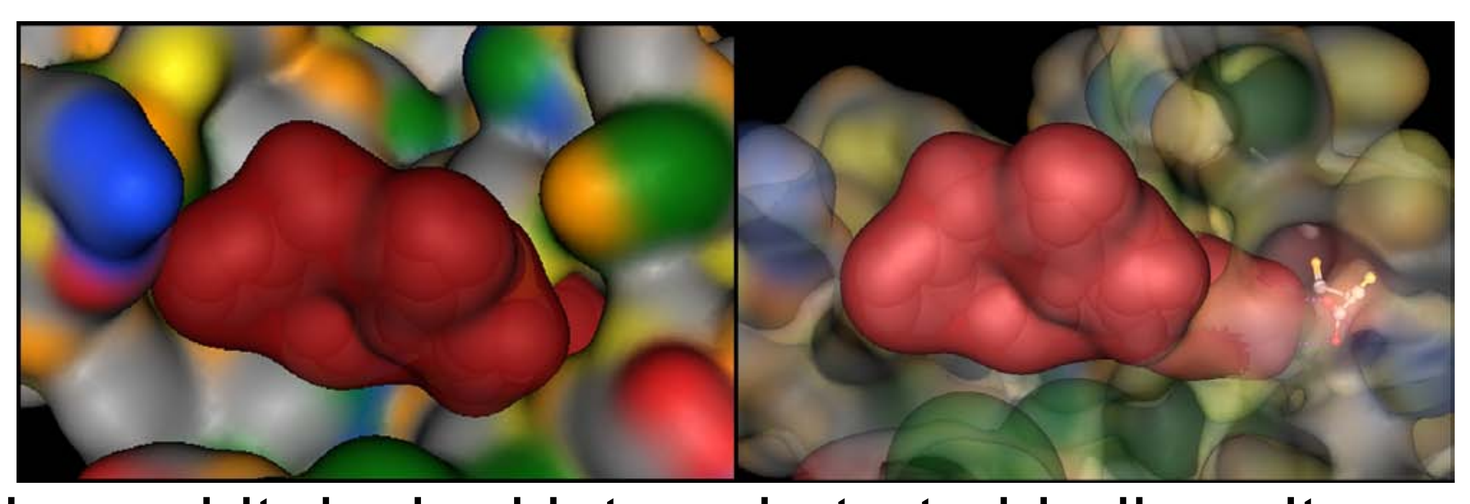

Primary hit docked into substrate binding site
- Primary screen (~ 50 cpds): 3 hits, Ki ~ 1.5 μM
- Small optimization library (~ 30 cpds): Ki 245 nM

Successful Programs - You Have The Target, We Have Its MATCH™

Tranzyme's Motilin Receptor Antagonist Portfolio

Compound	Pharmacodynamics				PK/ADME		
	Binding	Antagonist	*Ex Vivo*	*In Vivo*	Rat, i.v. PK		
	K_i	IC_{50}	Rabbit Atrium	Strain Gauge	$t_{1/2}$	Clearance Cl	Bio. Avail. (F)
	nM	nM	pA_2	Dog i.v. (mg/kg)	minutes	ml/min/kg	
1	4.6	7.5	7.76	>0.3	146	57	<1
2	2.1	0.53	8.11	<0.3	139	33	4.5
3*	1.2	0.12	8.35	>0.3	24	45	13
4	10.3	5.4	7.8	>0.3	25	65	11.4

- Proof-of-concept achieved in preclinical dog CID model
 - Compound 3: efficacious at 2.5 & 7.5 mg/kg (dog, i.v.)
- Clinical candidate (TZP-201) selected

Selective Hits on Three Protein-Protein Interaction Targets

Target Number	Primary hits n/20K cpds	Confirmed Hits/Tested	Validated Hits/Tested	K_i in μM (# of cpds)
1	1948	82/192 (43%)	34/54 (64%)	<10 (10)
2	2375	85/86 (99%)	16/41 (39%)	<10 (5)
3	390	132/170 (78%)	2/56 (4%)	22 (2)

Highly Potent & Selective Primary Hits on Ion Channels

Hit Number	Primary Target IC_{50} (μM)	Liability Target IC_{50} (μM)	Fold Selectivity
1	0.11	42	382
2	0.02	5	250
3	0.21	>30	>142
4	0.39	>30	>77
5	0.79	21	27

Motilin Receptor Agonists

Motilin Agonist (EC_{50} = 39 nM)

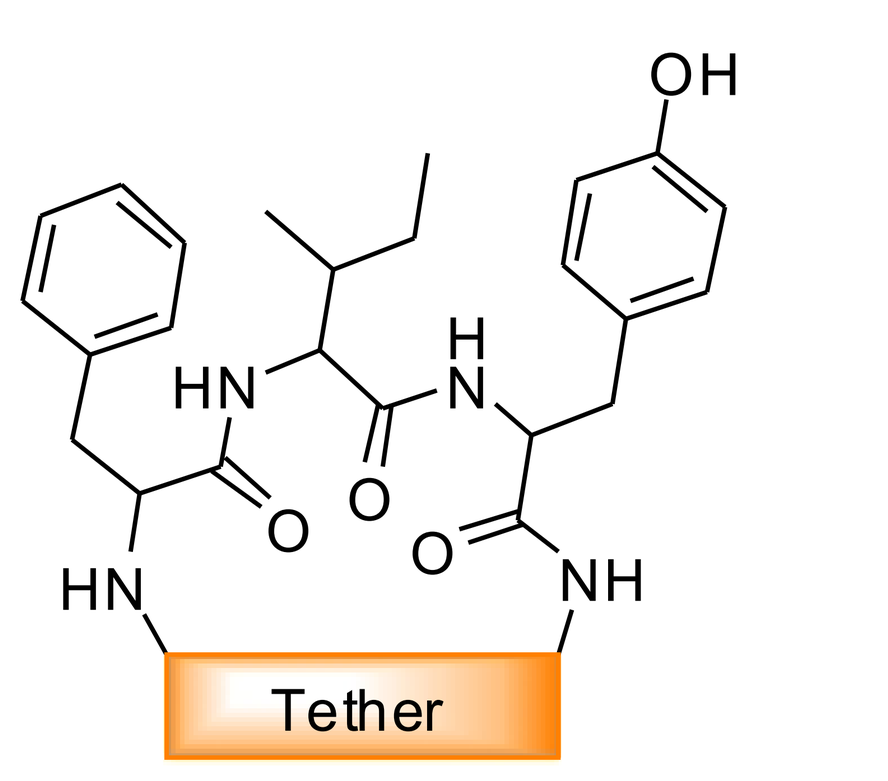
OH
HN
H
N
O
HN
NH
Tether

- Design based on key elements of motilin pharmacophores
 - Phe[1], Ile[4], Tyr[7]
- One library designed (n ~30 molecules)
 - Constant amino acid sequence
 - Variable macrocyclic ring size
- Hit potentcy dependent on macrocyclic ring size:
 - 16 atoms: K_i = 106 nM
 - 17 & 18 atoms: K_i = 2 mM